A Phase I/II, dose escalation study to assess the safety and tolerability of VAL201 in patients with locally advanced or metastatic prostate cancer and other advanced solid tumours

Patient characteristics

Recruitment Criteria:
- Patients with incurable, locally advanced or metastatic prostate cancer where a policy of intermittent hormone therapy has been decided
- Rising PSA on three samples; each over 2 weeks apart, with the last two values being greater than 2 ng/mL, at least 25% over the nadir.
- Absent or very mild prostate cancer-related symptoms.
- No plans for any therapy for prostate cancer in the next two months

Patients were retained on trial for up to 6 x 3 week cycles, unless removed earlier due to:
- Patient/clinician decision
- Adverse events (whether or not related to the treatment)
- Progressive disease
- Other treatment options became available to them outside of the trial

If removed for reasons other than progressive disease, progression was assessed at the final dosing visit by PCWG2 criteria

Parameter	Mean	Range
Age at enrolment (years)	72.4	60 – 84
Weight (kg)	82.62	64.8 – 96.5
BMI (kg/m2)	26.77	23.14 – 30.17
Time since diagnosis (years)	9.76	4.7 – 17.6

Dose	Number of patients	Dose-limiting toxicity	Patients completing 6 cycles	Patients with no progressive disease
0.5 mg/kg	1	0	1 (100%)	1 (100%)
1 mg/kg	1	0	1 (100%)	0
2 mg/kg	3	0	0	1 (33%)
4 mg/kg	5*	0	2 (40%)	3 (60%)
8 mg/kg	1	1	0	1 (100%)
TOTAL	11	1	4 (36%)	6 (55%)
*Excludes 1 patient on 4 mg/kg that received only one dose				

Subject	Cancer Type	VAL201 Dose	Date of diagnosis	Stage at Screening	T	N	M	PCWG2 Responder	6 Cycles Completed
101001	Prostate Cancer	0.5 mg/kg	NOV2009	Metastatic	x	x	Ib	Y	Y
101003	Prostate Cancer	1 mg/kg	APR2007	Locally Advanced	2b	0	0	-	Y
101005	Prostate Cancer	2 mg/kg	JUN2007	Metastatic	3b	1	1c	-	-
101006	Prostate Cancer	2 mg/kg	1998	Locally Advanced	x	x	x	-	-
101007	Prostate Cancer	2 mg/kg	NOV2006	Locally Advanced	3a	0	0	Y	-
101008	Prostate Cancer	4 mg/kg	2007	Metastatic	0	0	1	Y	-
101009	Prostate Cancer	4 mg/kg	JUL1998	Locally Advanced	2c	0	0	-	-
101010	Prostate Cancer	4 mg/kg	MAR2008	Locally Advanced	3	0	0	-	-
101011	Prostate Cancer	4 mg/kg	MAR2011	Locally Advanced	3b	1	0	Y	Y
101012	Prostate Cancer	4 mg/kg	JUL2012	Metastatic	3b	1	1	Y	-
101013	Prostate Cancer	8 mg/kg	2009	Locally Advanced	3a	0	0	Y	-
101014	Prostate Cancer	4 mg/kg	FEB2003	Metastatic	X	0	1C	-	Y

A Phase I/II, dose escalation study to assess the safety and tolerability of VAL201 in patients with locally advanced or metastatic prostate cancer and other advanced solid tumours

Disease Impact

Patients were defined as responding to the treatment by assessment using PCWG2 criteria. PCWG2 was assessed at regular time points during the trial. If no progressive disease was declared at any time that the patient was on the trial, the patient is declared a responder.
To meet PCWG2 criteria for non-progression (stable disease), patients must observe:

- PSA increase no greater than 25%
- Tumour size increase no greater than 20%
- No more than 1 additional metastatic lesion

Disease impact can be considered by the PSA biomarker, with the level of increase in PSA providing a surrogate measure for the level of increase in cancerous activity; and by direct measurement of tumour sizes (primary or metastatic) by MRI or CT scan.

Subject	VAL201 Dose	Screening scan tumour location	Tumour size	Final cycle scan tumour location	Tumour size
101001	0.5 mg/kg	Seminal Vesicle	28 mm	Seminal Vesicle	28 mm
101003	1 mg/kg	None measurable		None measurable	
101005	2 mg/kg	None measurable		None measurable	
101006	2 mg/kg	None measurable		None measurable	
101007	2 mg/kg	None measurable		None measurable	
101008	4 mg/kg	None measurable		None measurable	
101009	4 mg/kg	None measurable		None measurable	
101010	4 mg/kg	None measurable		None measurable	
101011	4 mg/kg	None measurable		None measurable	
101012	4 mg/kg	None measurable		None measurable	
101013	8 mg/kg	Mesorectal Space	49 mm	Mesorectal Space	55 mm
101014	4 mg/kg	Lung Lesion 1	22 mm	Lung Lesion 1	20 mm
		Lung Lesion 2	17 mm	Lung Lesion 2	16 mm

Subject	VAL201 Dose	Pre-treatment PSA doubling time (days)	Post-treatment PSA doubling time (days)	Maximal PSA decline
101001	0.5 mg/kg	68.4	638	N/A
101003	1 mg/kg	623.1	191.2	N/A
101005	2 mg/kg	26.6	59.6	N/A
101006	2 mg/kg	28.3	48.8	N/A
101007	2 mg/kg	298.6	336.9	N/A
101008	4 mg/kg	36.8	N/A*	50%
101009	4 mg/kg	87.4	310.5	N/A
101011	4 mg/kg	455.9	7965	22%**
101012	4 mg/kg	118.9	281.7	N/A
101013	8 mg/kg	7.9	205.7	N/A
101014	4 mg/kg	154.0	387.3	N/A
Mean* **		**186.91**	**1030.67**	

*Doubling time is "infinite" as PSA was consistently decreasing, excluded from Mean

**PSA decreased initially, but overall high doubling time shows it levelled at the baseline screening level

***Post treatment doubling time is higher than pre-treatment doubling time, with statistical significance of $p<0.05$ using 2-tailed Wilcoxon Signed-Rank Test for paired samples

2

A Phase I/II, dose escalation study to assess the safety and tolerability of VAL201 in patients with locally advanced or metastatic prostate cancer and other advanced solid tumours

Safety and Tolerability

VAL201 was well tolerated with the single dose-limiting toxicity being hypertension in the 8 mg/kg group in a patient with pre-existing hypertension. The patient received additional treatment for their hypertension and continued on the trial at the same dose of VAL201.
No Maximum Tolerated Dose was established, and all dose levels remain available for future testing if required.

Pharmacokinetic profiles were obtained from only the final two patients, with increased dose showing increased exposure as expected. These patients were the only patients to be dosed after a protocol amendment which enabled a more sensitive analytical procedure to be implemented.
Levels of patient exposure to VAL201 showed it to be well absorbed following the subcutaneous administration.
The pharmacokinetic data shown as a mean value of at least 3 separate blood collection profiles for each patient with error bars showing the Standard Error.

Event	0.5 mg/kg (N=1)	1 mg/kg (N=1)	2 mg/kg (N=3)	4 mg/kg (N=6)	8 mg/kg (N=1)	Total (N=12)
	n (%)	n (%)	n (%)	n (%)	n (%)	n (%)
General disorders and administration site conditions	**1 (100.0)**	**1 (100.0)**	**3 (100.0)**	**5 (83.3)**	**1 (100.0)**	**11 (91.7)**
Fatigue	0	0	3 (100.0)	2 (33.3)	0	5 (41.7)
Injection site rash	0	0	0	5 (83.3)	0	5 (41.7)
Injection site reaction	1 (100.0)	1 (100.0)	2 (66.7)	1 (16.7)	0	5 (41.7)
Injection site pain	0	0	0	2 (33.3)	1 (100.0)	3 (25.0)
Injection site erythema	0	0	0	1 (16.7)	1 (100.0)	2 (16.7)
Gastrointestinal disorders	**0**	**0**	**0**	**1 (16.7)**	**0**	**1 (8.3)**
Dyspepsia	0	0	0	1 (16.7)	0	1 (8.3)
Musculoskeletal and connective tissue disorders	0	0	0	1 (16.7)	0	1 (8.3)
Muscle Spasm	0	0	0	1 (16.7)	0	1 (8.3)
Vascular disorders	0	0	0	1 (16.7)	1 (100.0)	2 (16.7)
Hypertension	0	0	0	1 (16.7)	1 (100.0)	2 (16.7)
Cardiac disorders	0	0	0	1 (16.7)	0	1 (8.3)
Bradycardia	0	0	0	1 (16.7)	0	1 (8.3)

	Patient 101013	Patient 101014
Dose	8 mg/kg	4 mg/kg
t_{max}	31 mins	40 mins
C_{max}	3323 ng/ml	2205 ng/ml
$t_{1/2}$	59 min	40 min
$AUC_{0\text{-}inf}$	5.0 ug/ml*h	3.8 ug/ml*h

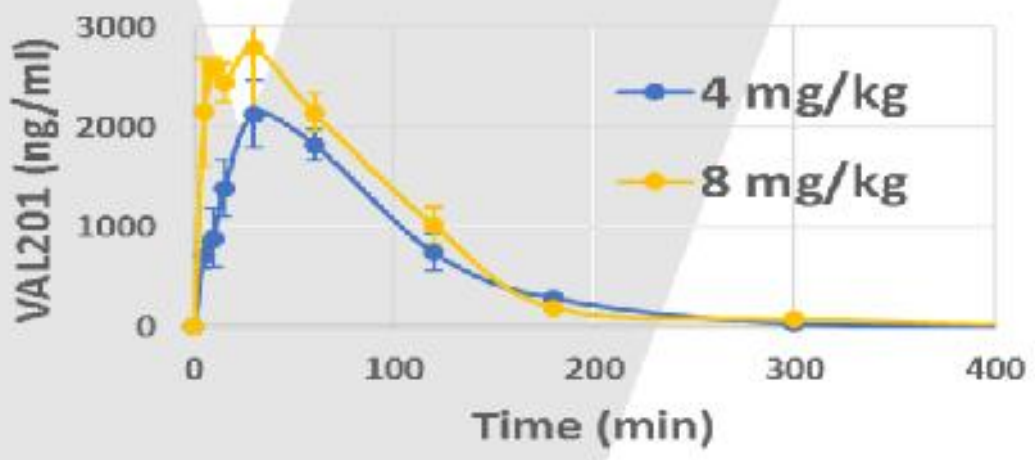

VAL201 (ng/ml)
3000
2000
1000
0
0
100
200
300
400
Time (min)
4 mg/kg
8 mg/kg

3